EEG decoding reveals a link between visual working memory fidelity and the magnitude of similarity-induced memory bias

Nursima Ünver[1,2,3], Rosanne L. Rademaker[2], & Keisuke Fukuda[1,3]

[1]University of Toronto, [2]Ernst Strüngmann Institute for Neuroscience, [3]University of Toronto Mississauga

Background

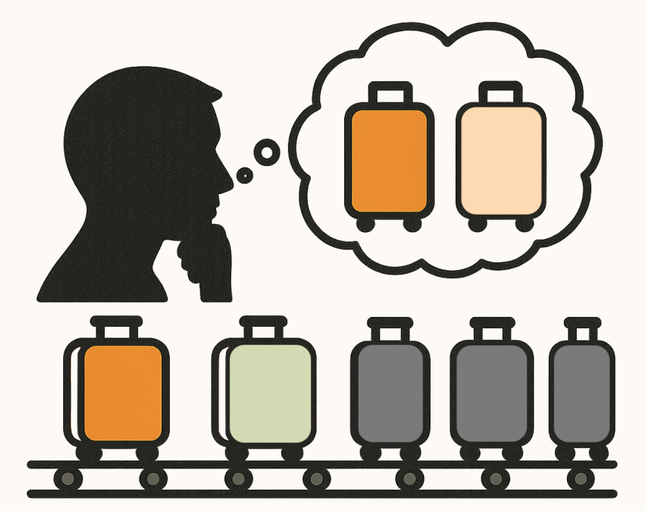

Visual working memory (VWM) reports can be **biased toward** novel visual input when VWM is **judged as similar** to input.

Similarity-induced Memory Bias *a.k.a.* SIMB.

How does target strength relate to SIMB?

Hypotheses

- Ünver et al., VSS 2023

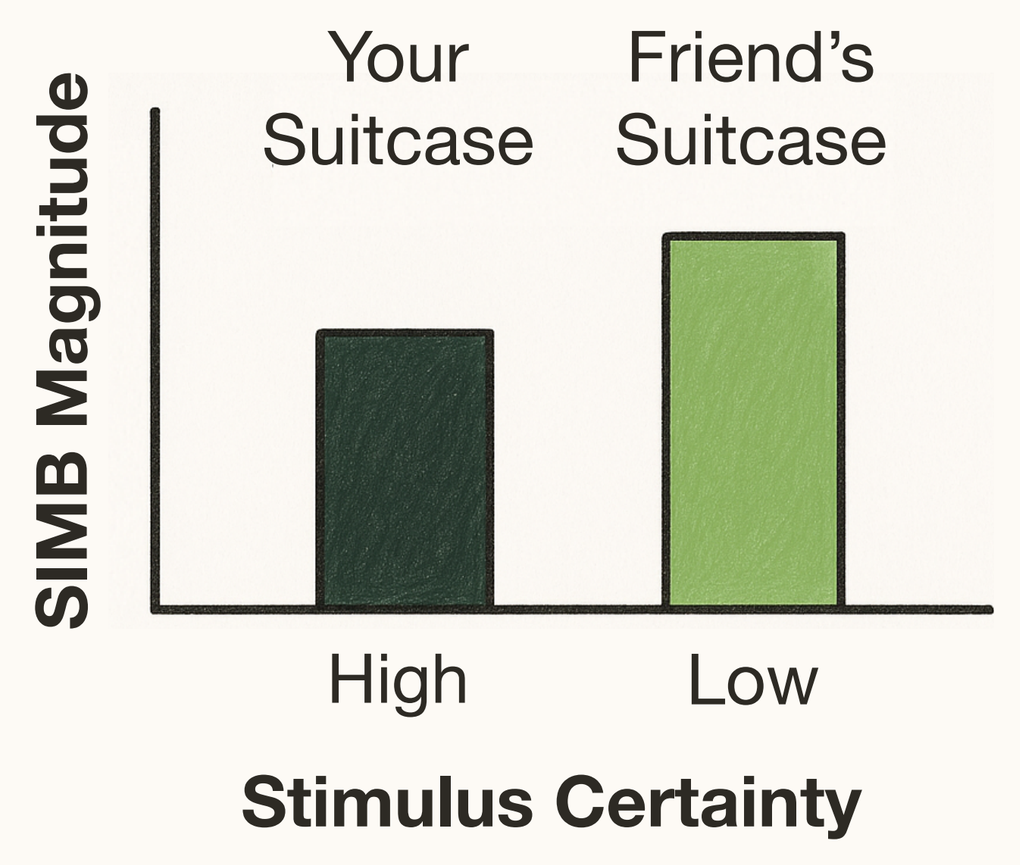

SIMB Magnitude
Your Suitcase
Friend's Suitcase
High
Low
Stimulus Certainty

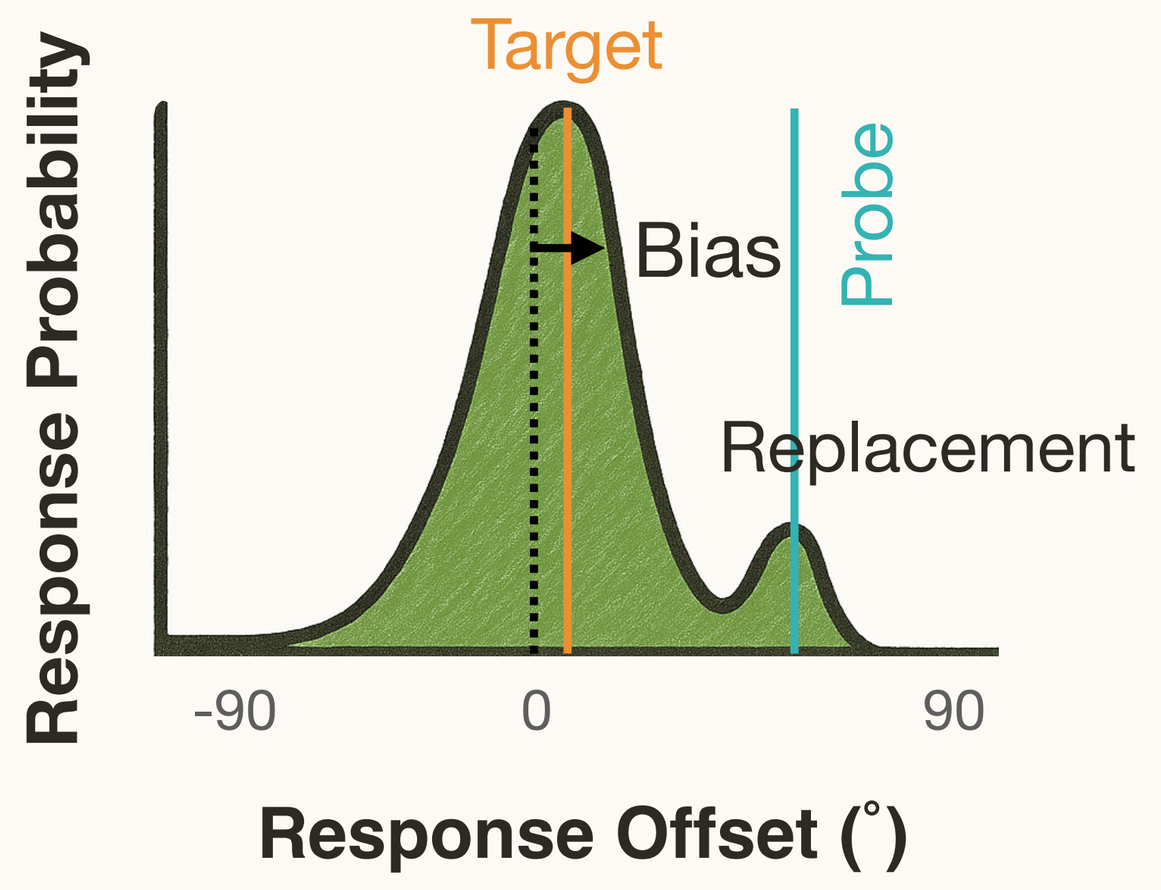

Response Probability
Target
Bias
Probe
Replacement
-90
0
90
Response Offset (˚)

- **Encoding Weakness Hypothesis:** SIMB stronger when the target is weakly encoded.
- **Probe Interference Hypothesis:** SIMB stronger when probe is more strongly encoded.

Method

Procedure

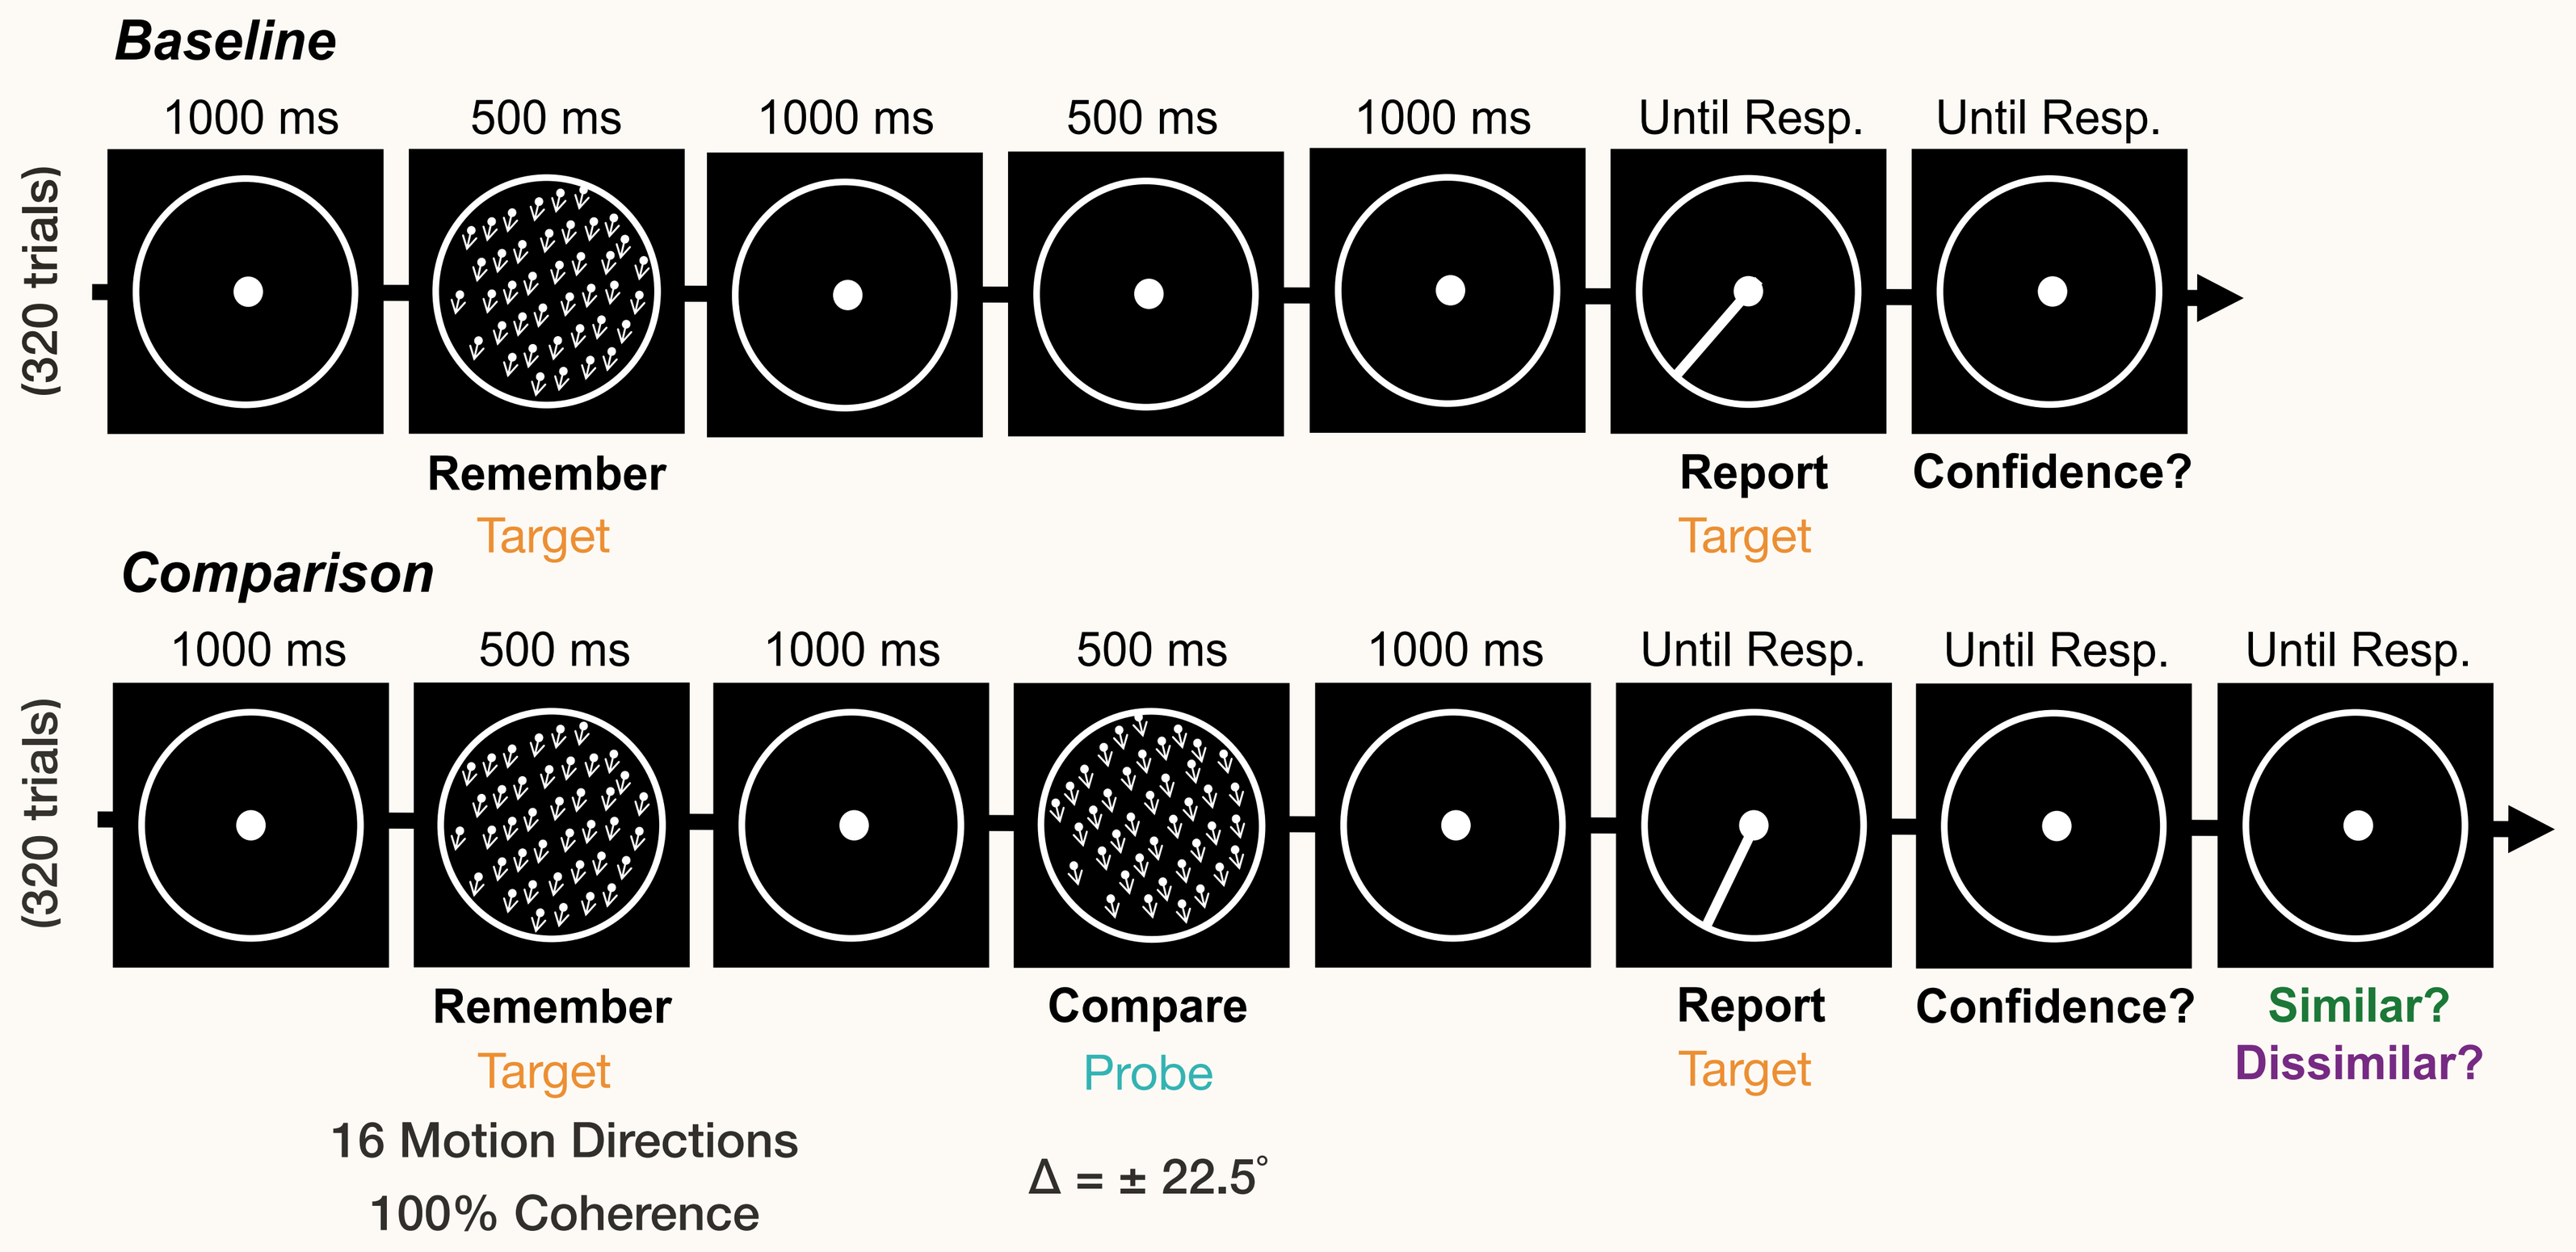

Baseline
(320 trials)
1000 ms
500 ms
1000 ms
500 ms
1000 ms
Until Resp.
Until Resp.
Remember
Target
Report
Target
Confidence?
Comparison
(320 trials)
1000 ms
500 ms
1000 ms
500 ms
1000 ms
Until Resp.
Until Resp.
Until Resp.
Remember
Target
Compare
Probe
Report
Target
Confidence?
Similar?
Dissimilar?
16 Motion Directions
100% Coherence
Δ = ± 22.5˚

Participants

N=23

Participants with ≥ 80 similar trials (N=19)

Participants with ≥ 80 dissimilar trials (N=17)

Trial-wise Median Split

For each participant and target bin:

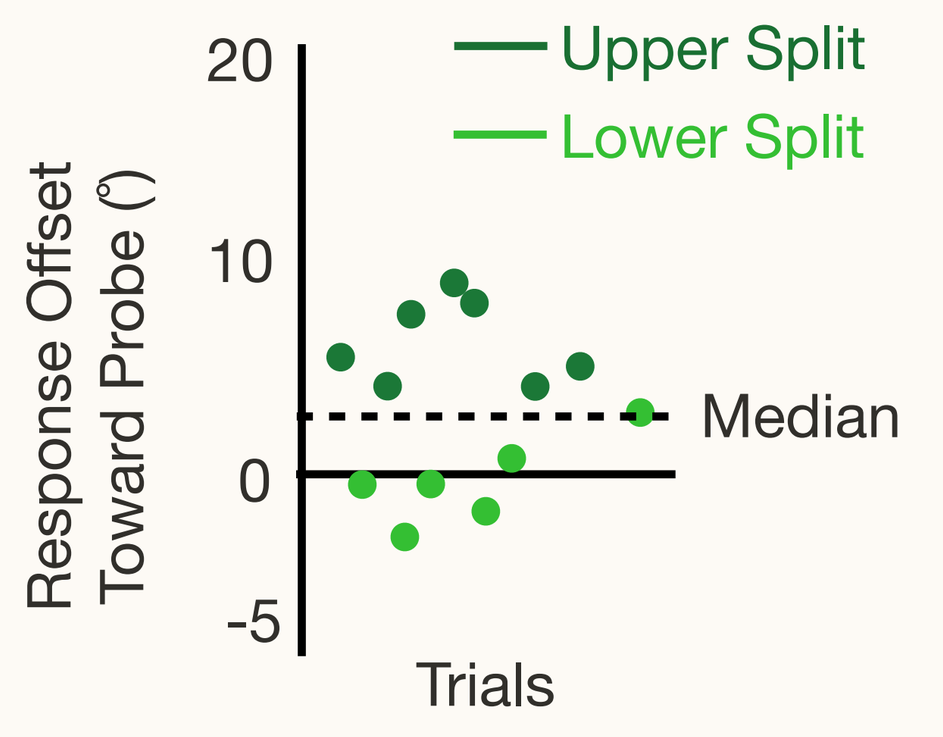

Response Offset Toward Probe (˚)
20
10
0
-5
Upper Split
Lower Split
Median
Trials

EEG Analyses

Decoding motion direction from posterior EEG channels

Trial-by-trial, within time point

Output = probability of the direction being the target

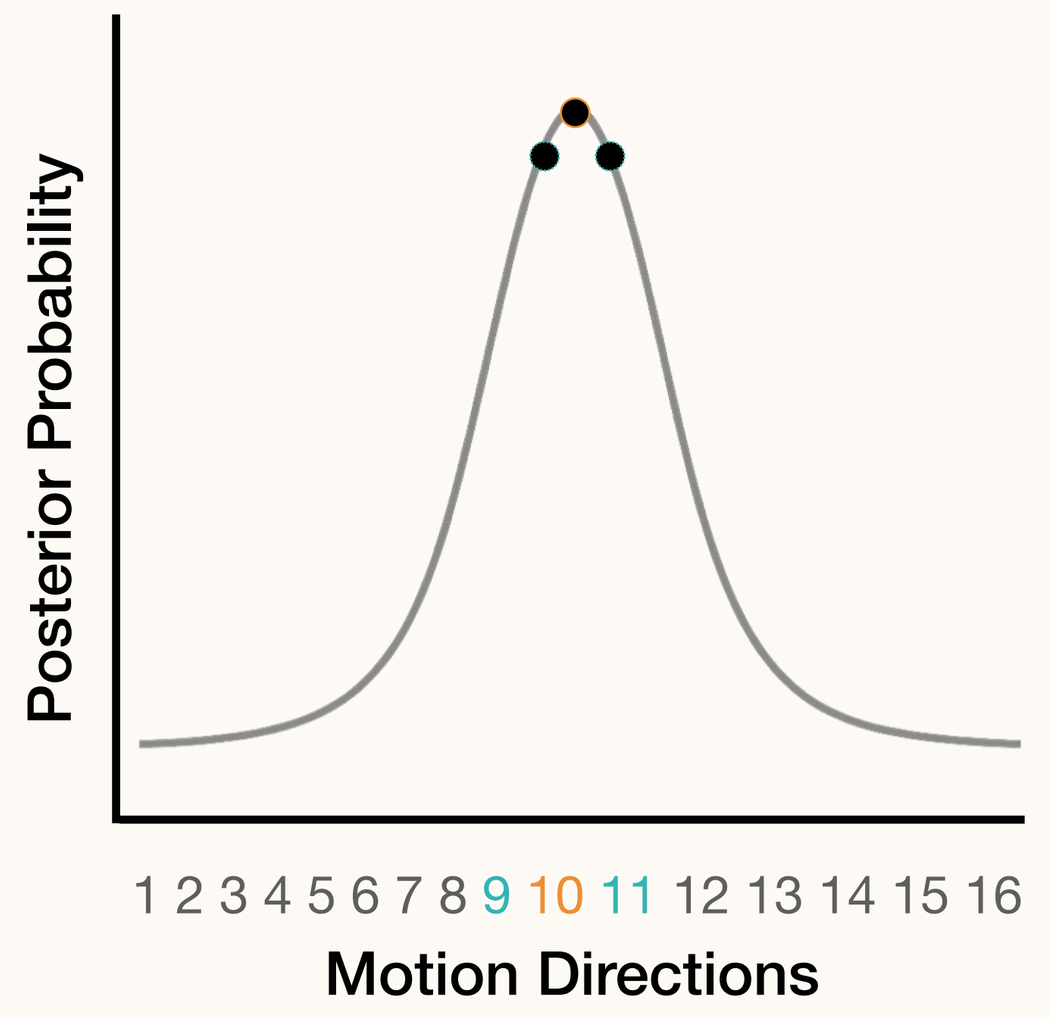

Posterior Probability
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16
Motion Directions

Behavioral Results

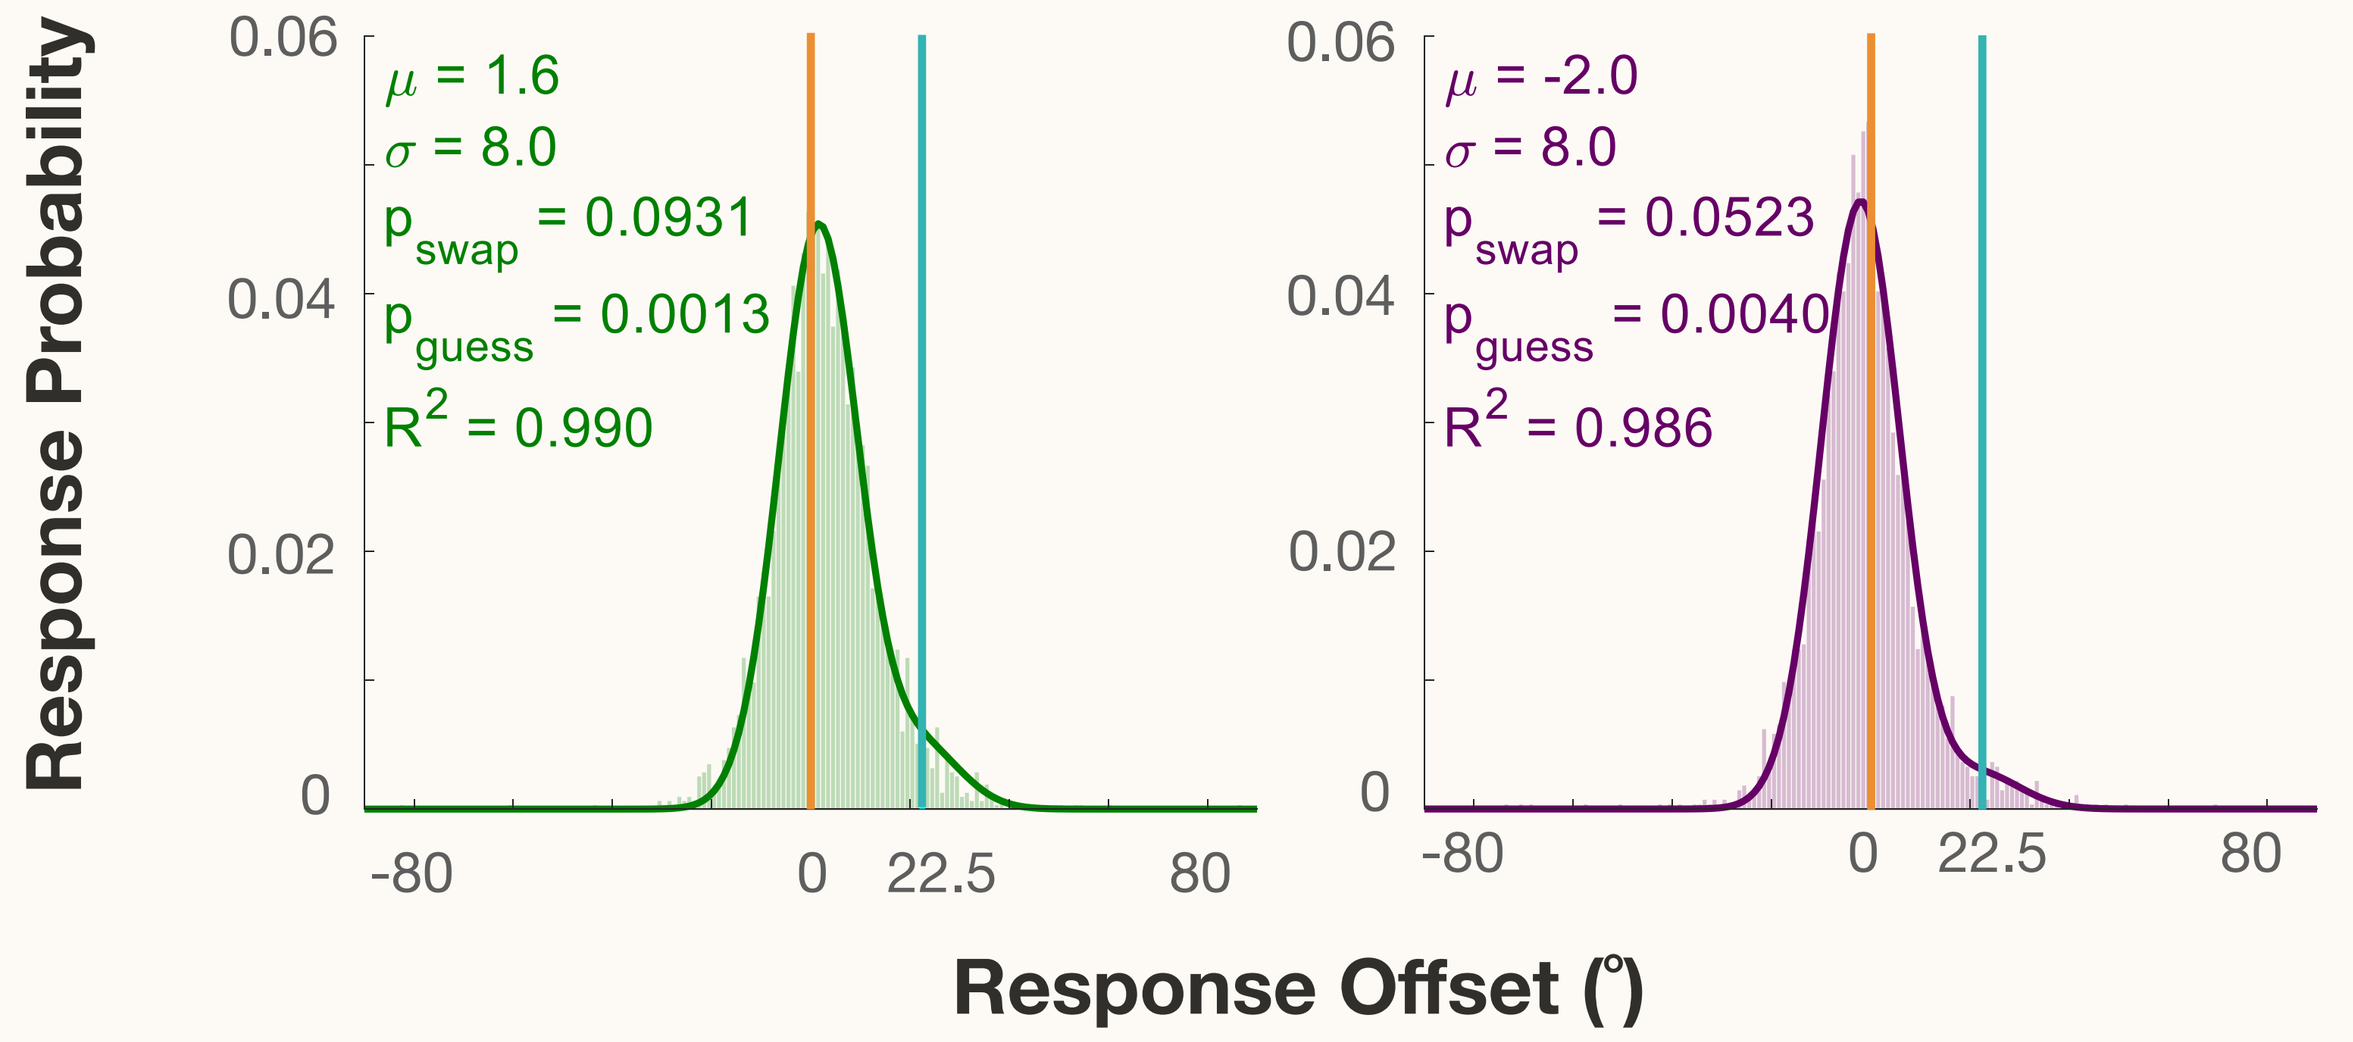

Response Probability
0.06
0.04
0.02
0
μ = 1.6
σ = 8.0
p swap = 0.0931
p guess = 0.0013
R2 = 0.990
μ = -2.0
σ = 8.0
p swap = 0.0523
p guess = 0.0040
R2 = 0.986
-80
0
22.5
80
Response Offset (˚)

Response offset shows **attraction toward similar** and **repulsion away from dissimilar** probe together with **replacement** errors!

Train on Baseline Trials (Decoder sees no probe)

- p<.05, cluster corrected

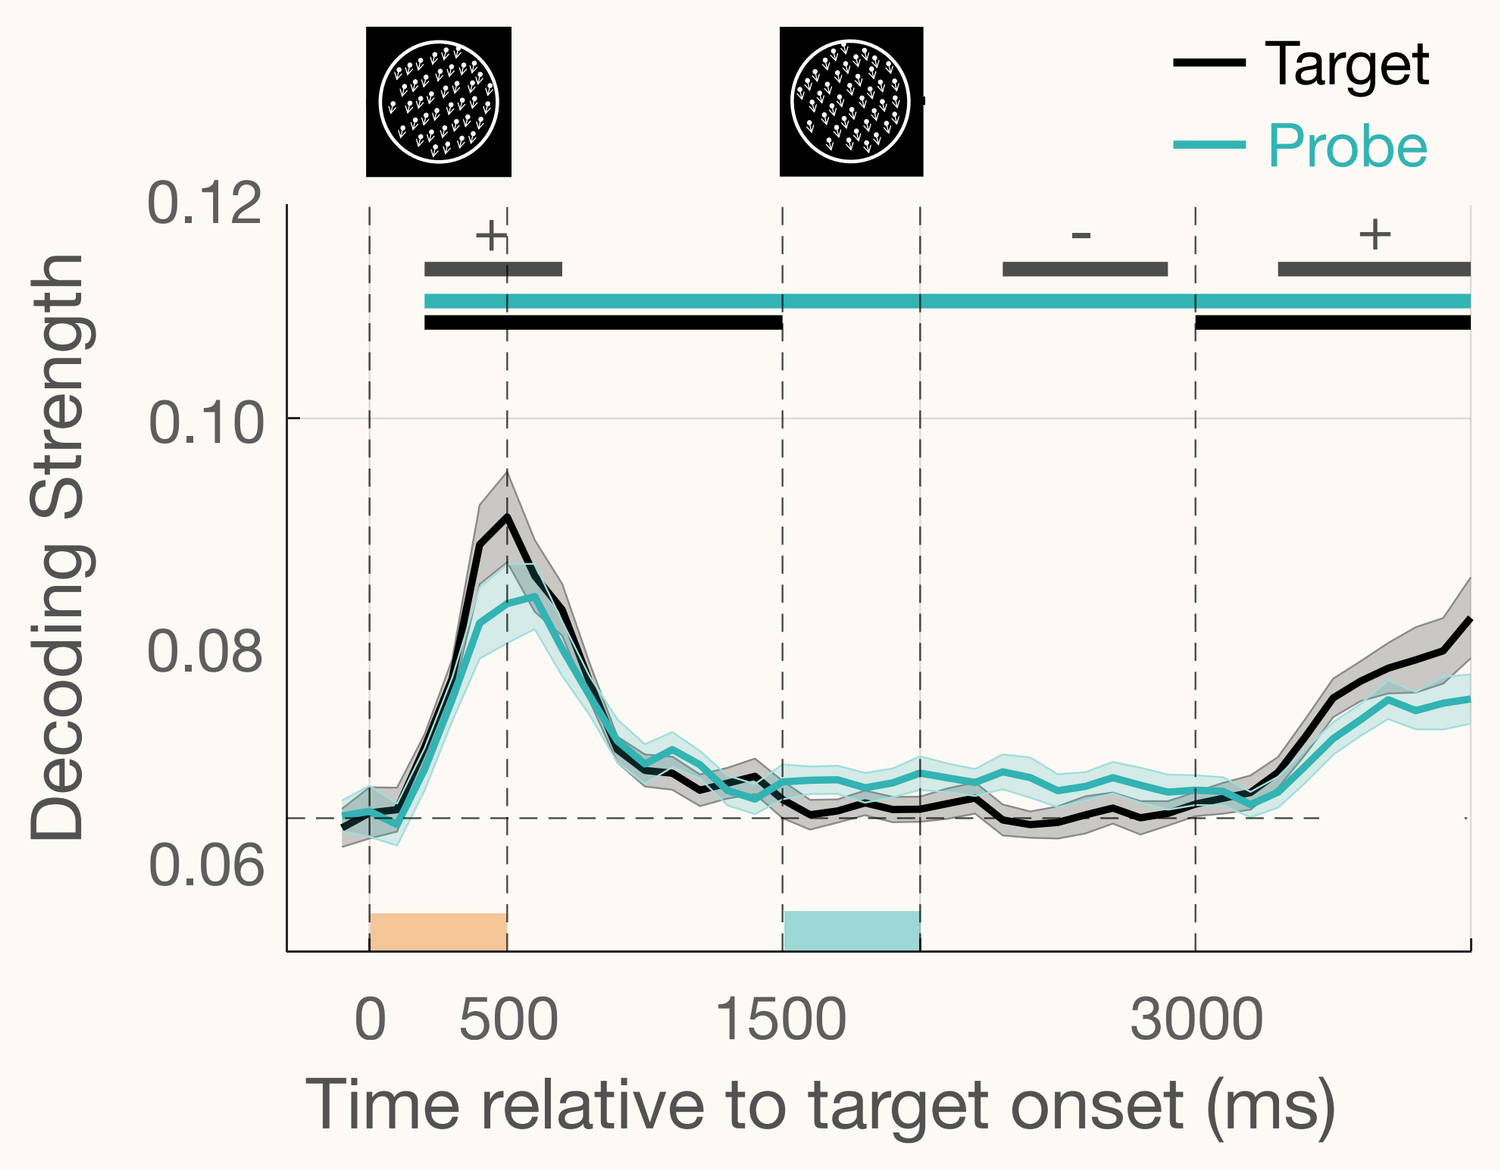

Target
Probe
Decoding Strength
0.12
0.10
0.08
0.06
0
500
1500
3000
Time relative to target onset (ms)

Is Target Encoding Strength Related to Behavioral Offset?

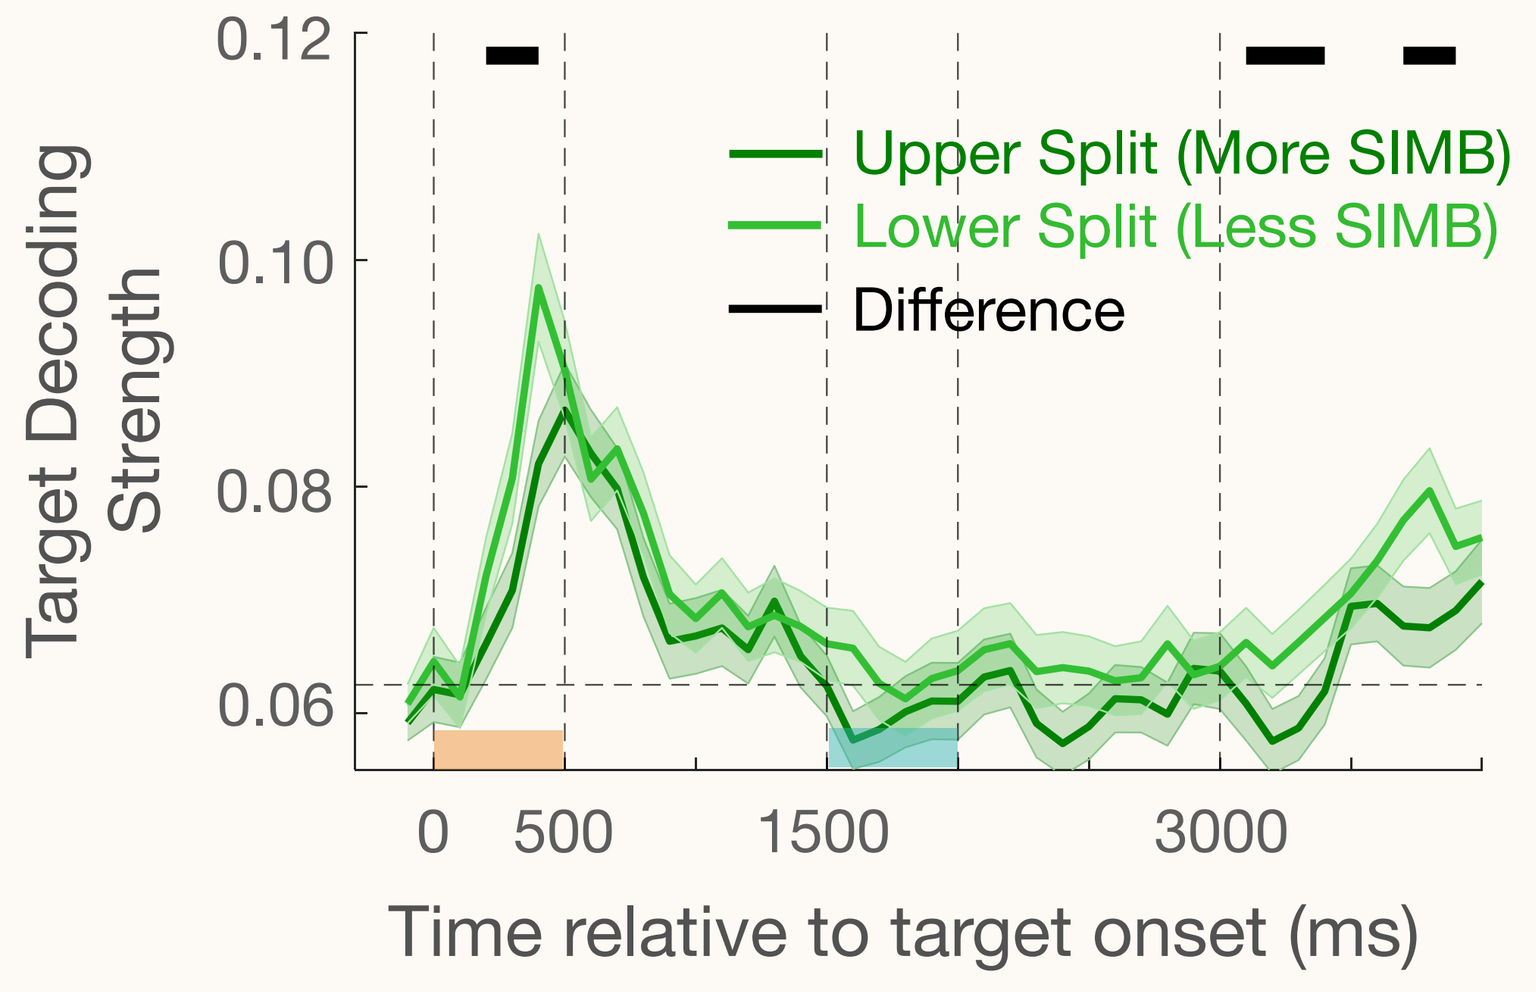

Target Decoding Strength
Upper Split (More SIMB)
Lower Split (Less SIMB)
Difference
0.12
0.10
0.08
0.06
0
500
1500
3000
Time relative to target onset (ms)

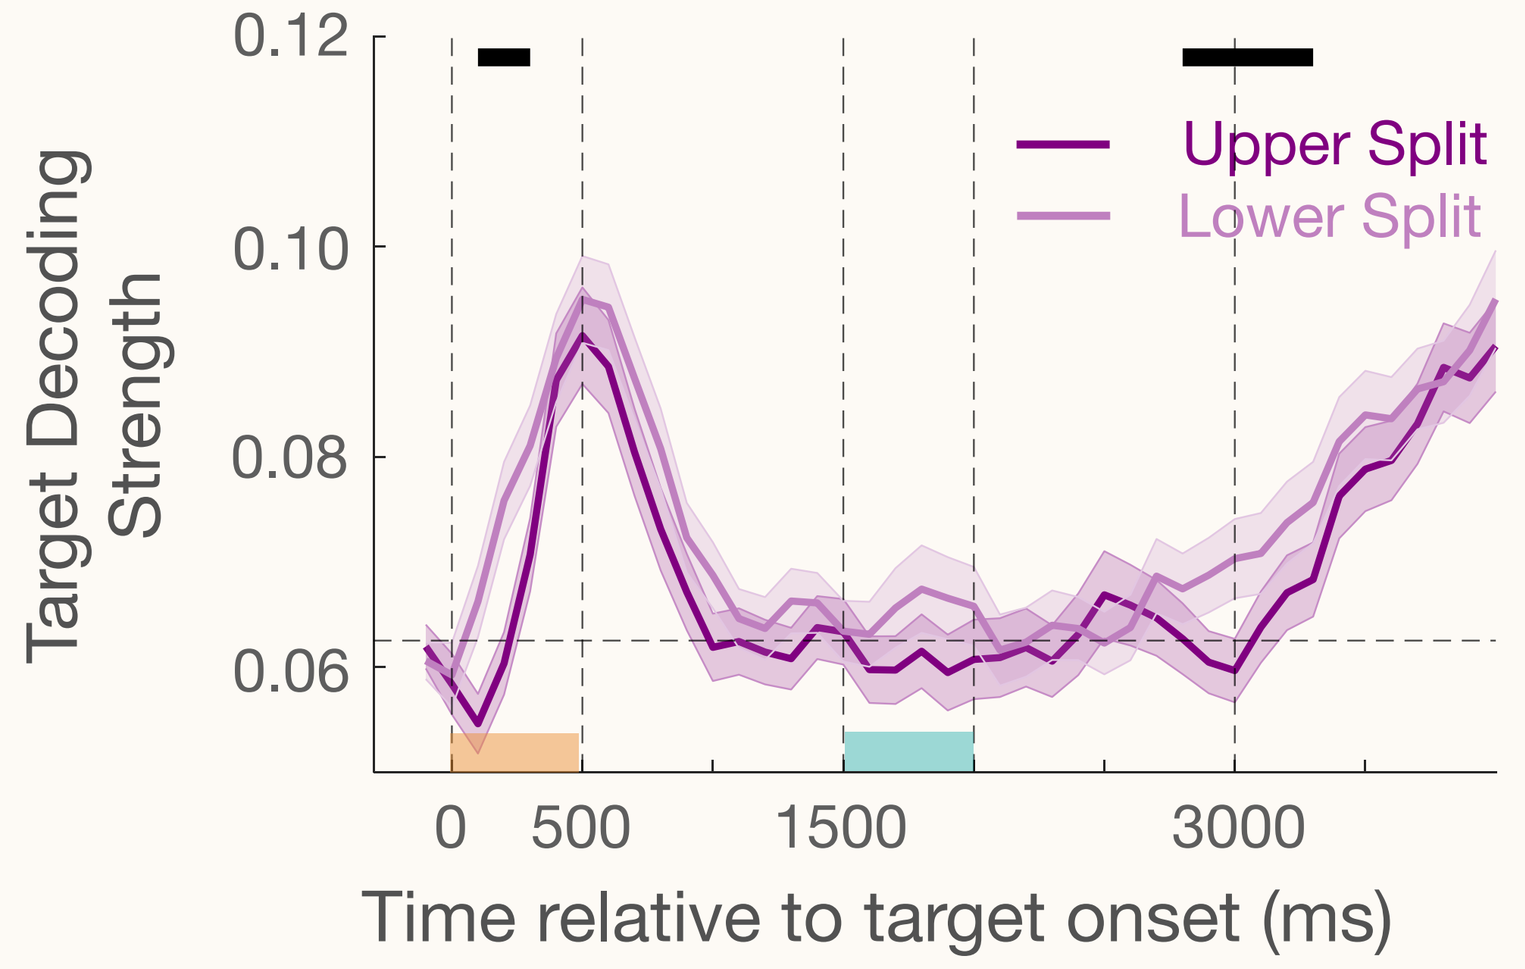

Target Decoding Strength
Upper Split
Lower Split
0.12
0.10
0.08
0.06
0
500
1500
3000
Time relative to target onset (ms)

SIMB is stronger (Upper Split) for more weakly encoded trials.

Is Probe Strength Related to Behavioral Offset?

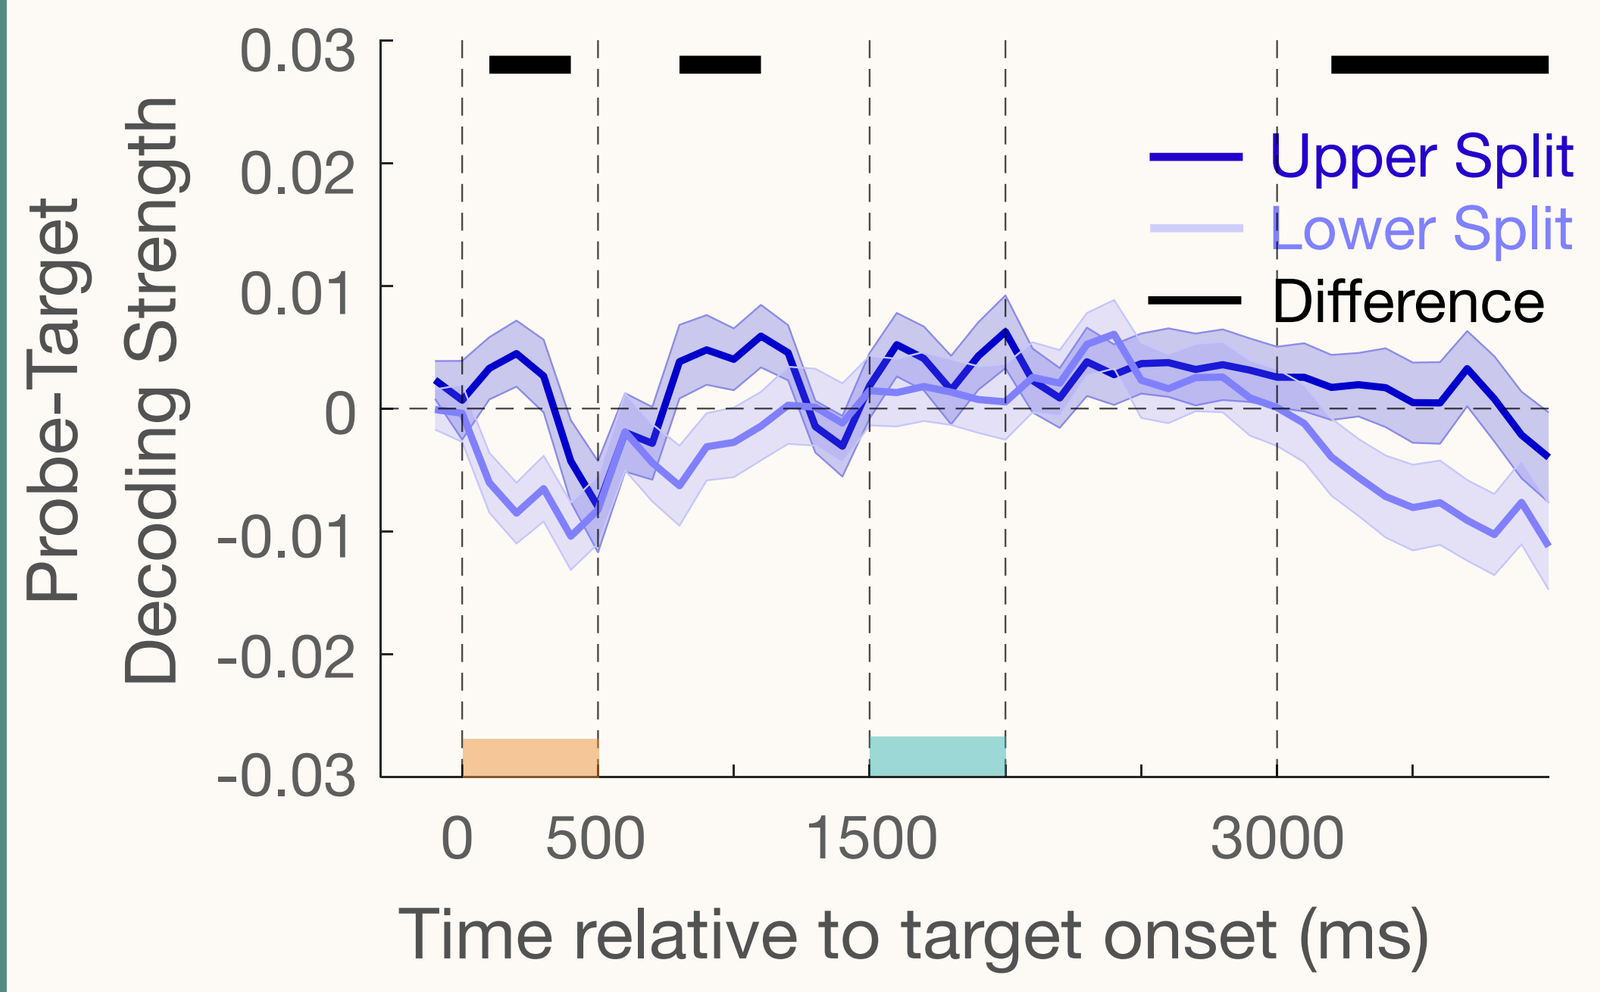

Probe-Target Decoding Strength
Upper Split
Lower Split
Difference
0.03
0.02
0.01
0
-0.01
-0.02
-0.03
0
500
1500
3000
Time relative to target onset (ms)

Trials with stronger bias have stronger probe evidence.

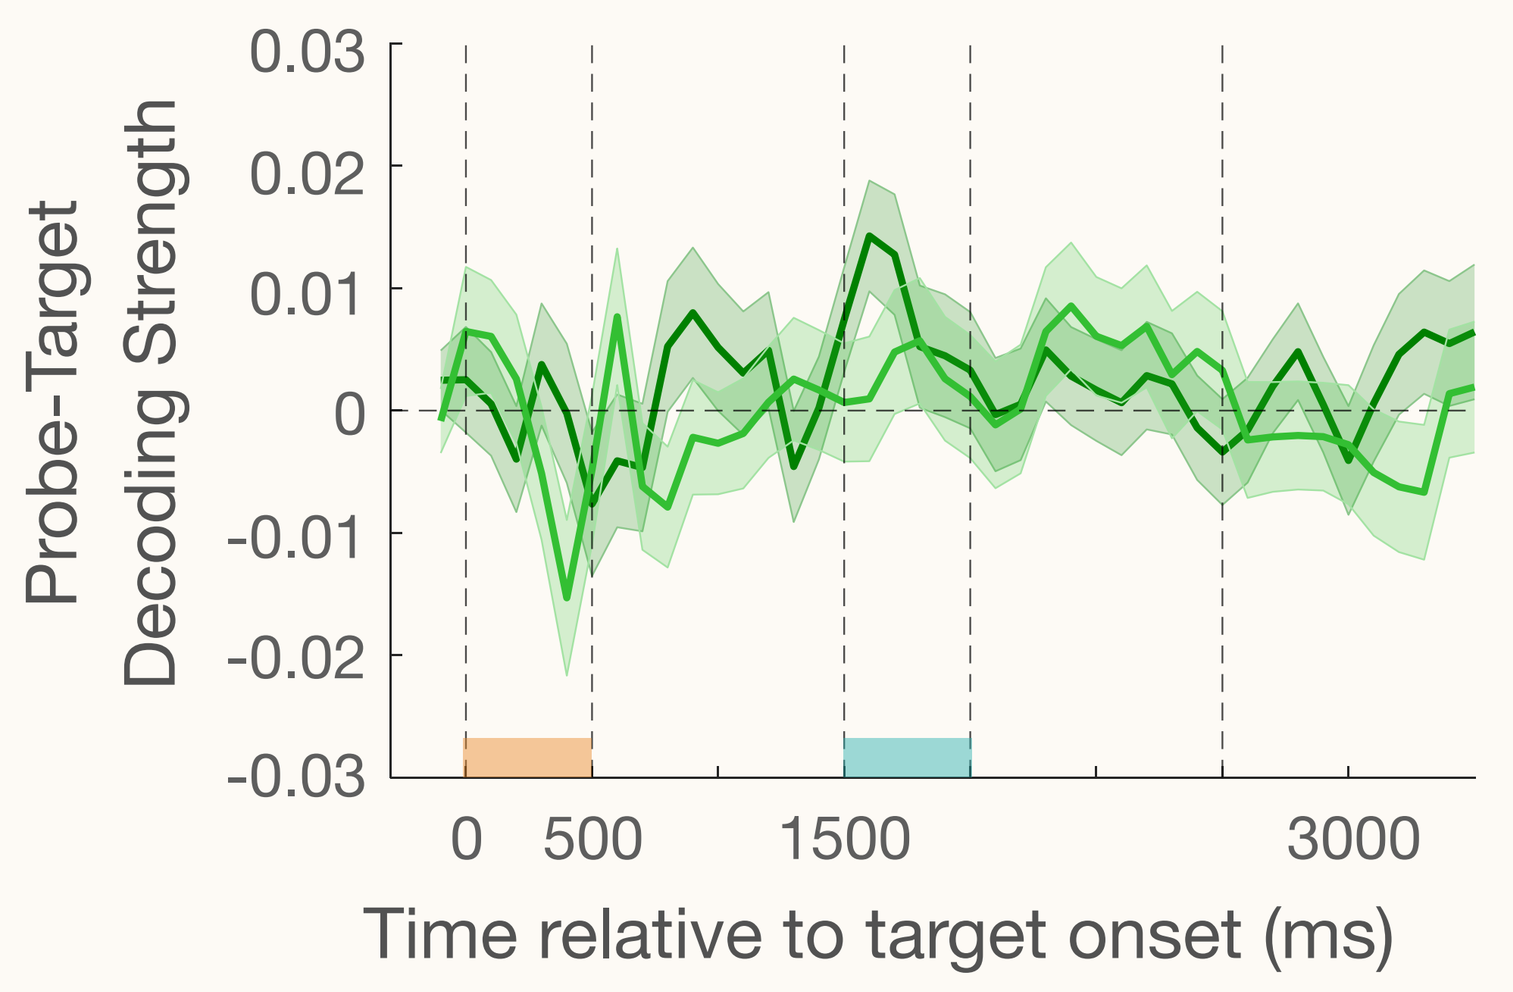

Probe-Target Decoding Strength
0.03
0.02
0.01
0
-0.01
-0.02
-0.03
0
500
1500
3000
Time relative to target onset (ms)

Probe evidence is not related to SIMB.

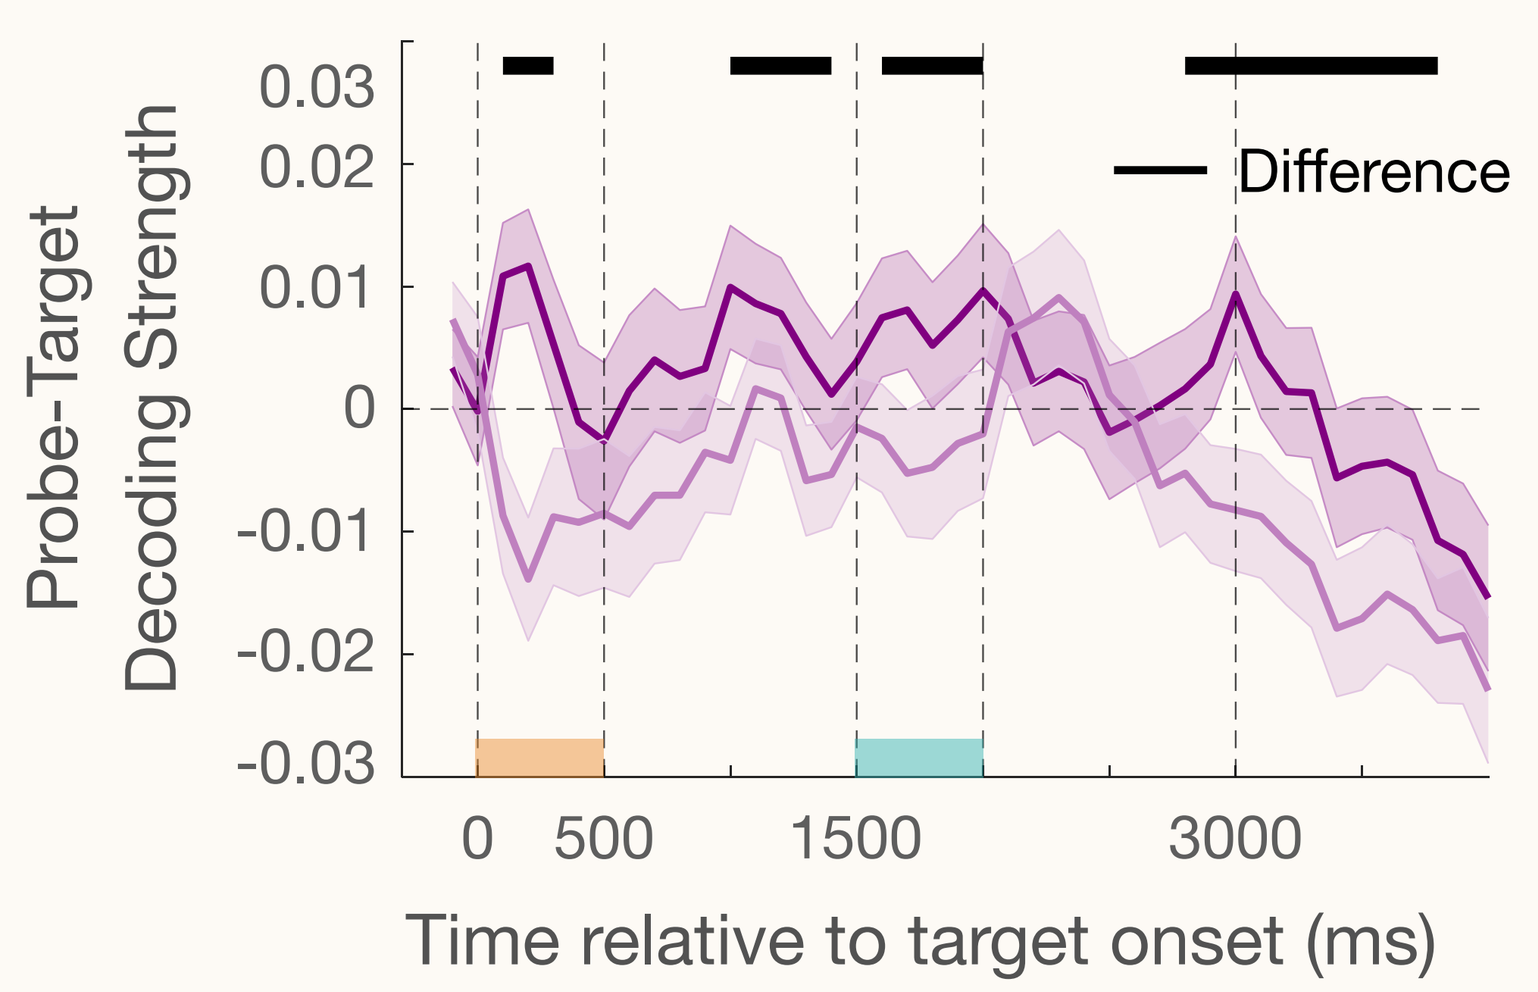

Probe-Target Decoding Strength
Difference
0.03
0.02
0.01
0
-0.01
-0.02
-0.03
0
500
1500
3000
Time relative to target onset (ms)

Stronger repulsion means less probe evidence.

Train on Comparison Trials (Decoder sees target and probe)

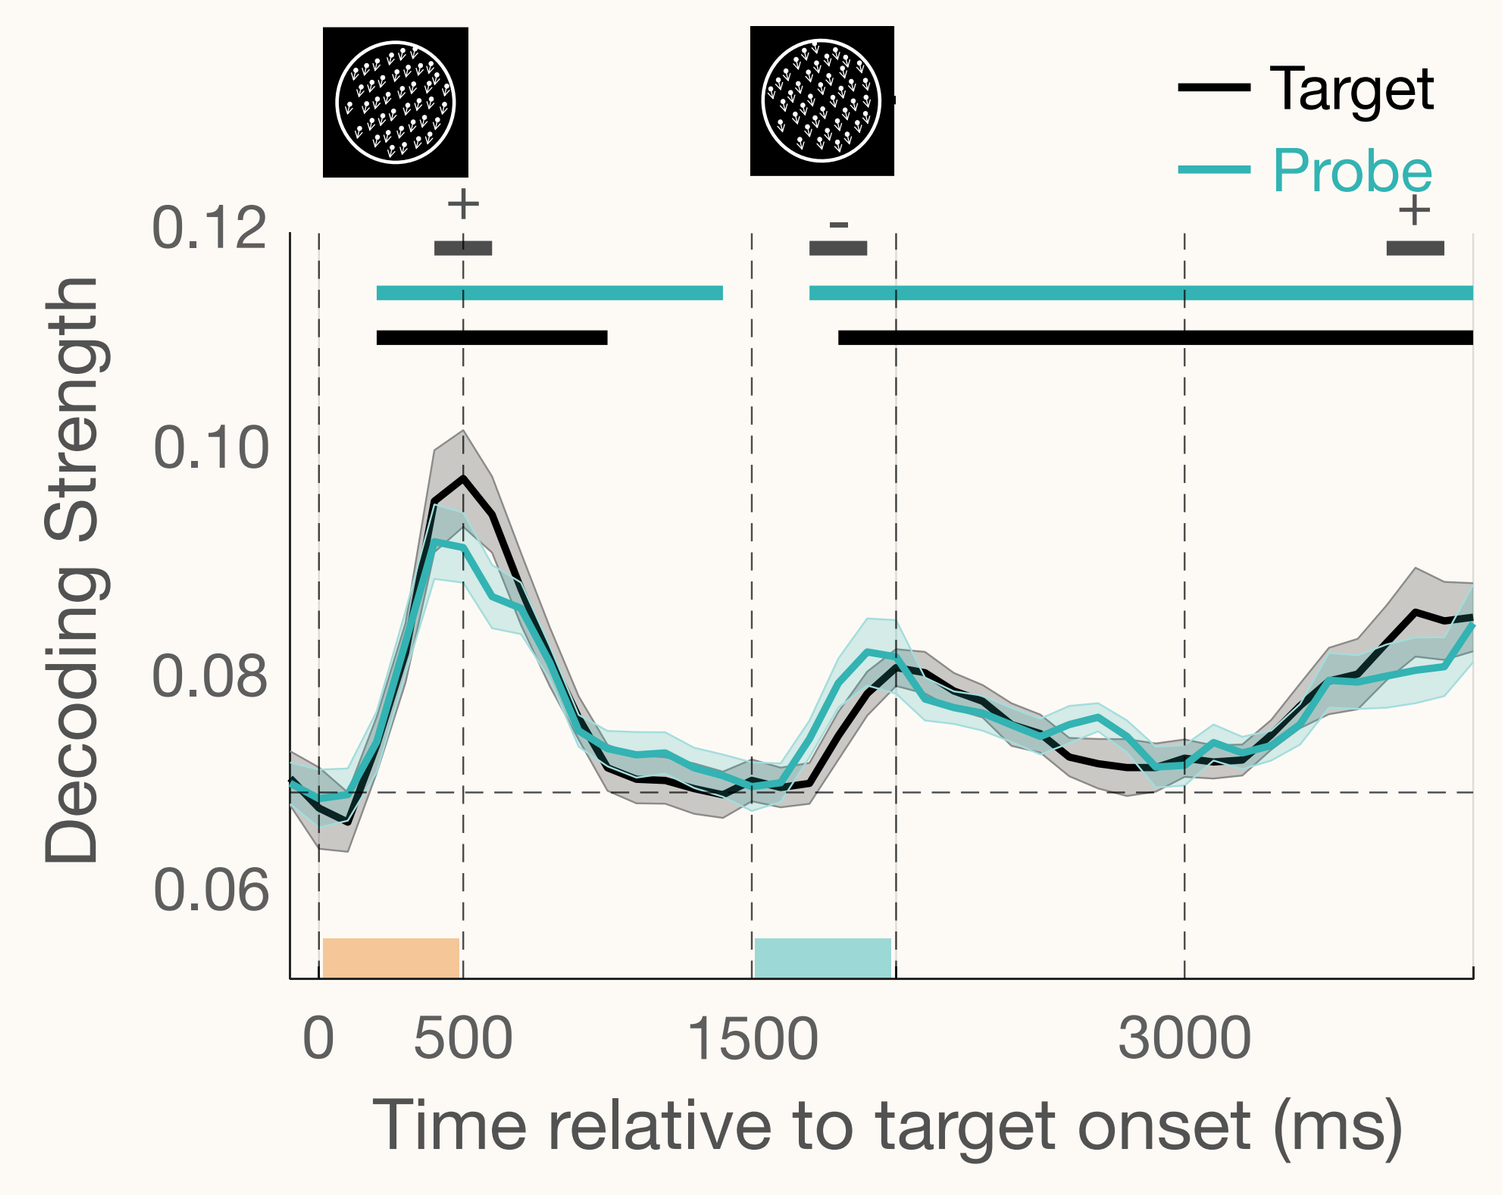

Target
Probe
Decoding Strength
0.12
0.10
0.08
0.06
0
500
1500
3000
Time relative to target onset (ms)

Is Target Encoding Strength Related to Behavioral Offset?

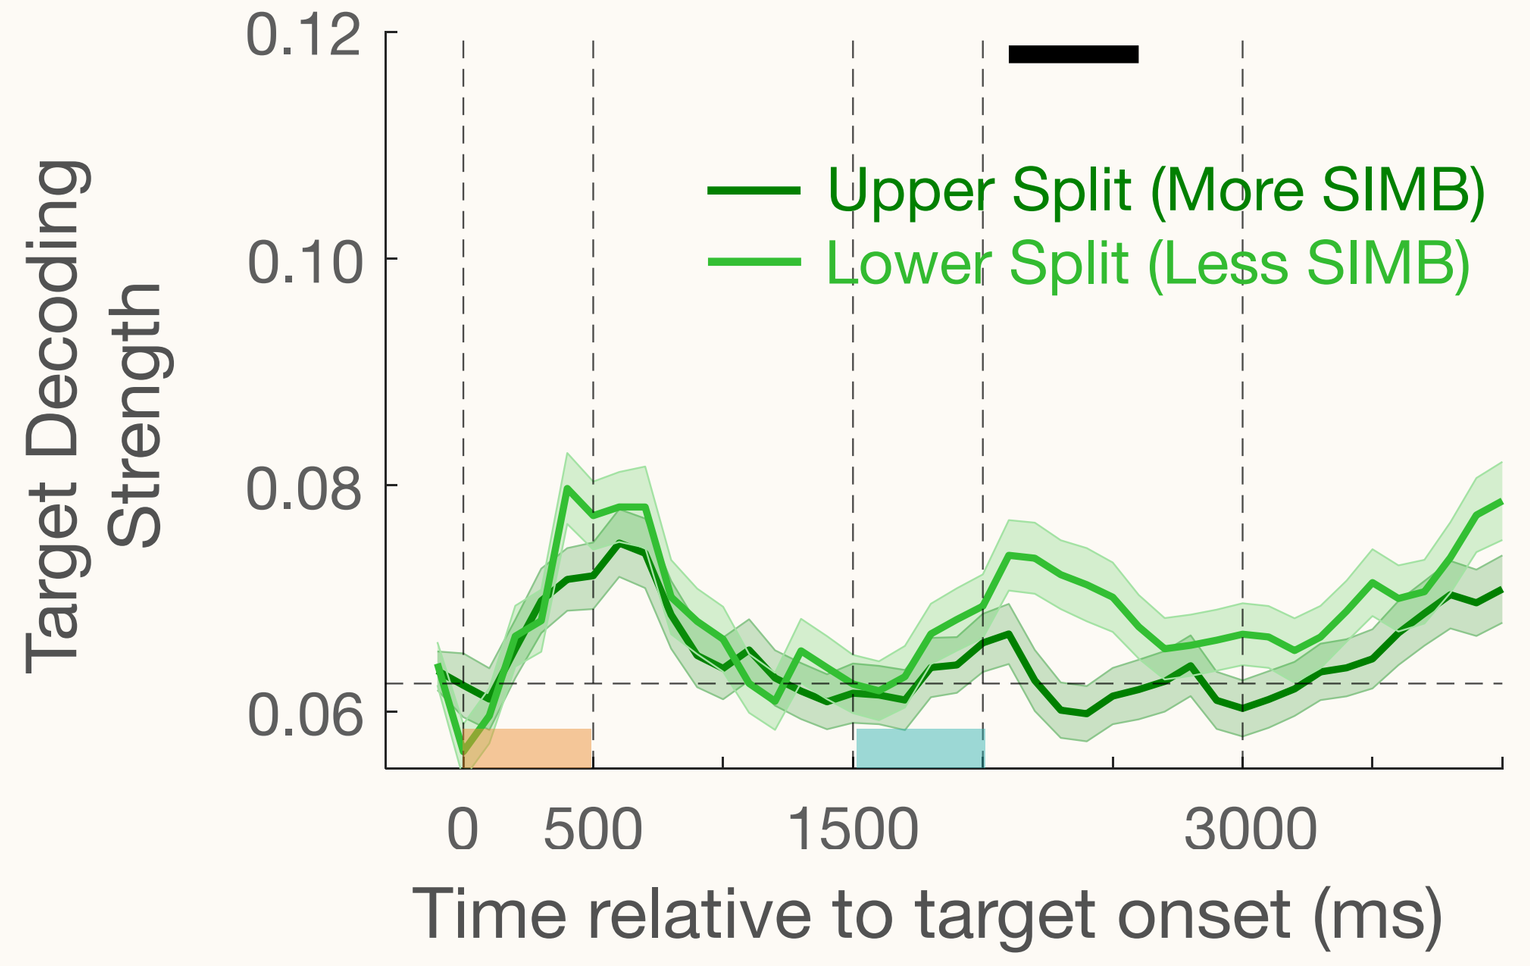

Target Decoding Strength
Upper Split (More SIMB)
Lower Split (Less SIMB)
0.12
0.10
0.08
0.06
0
500
1500
3000
Time relative to target onset (ms)

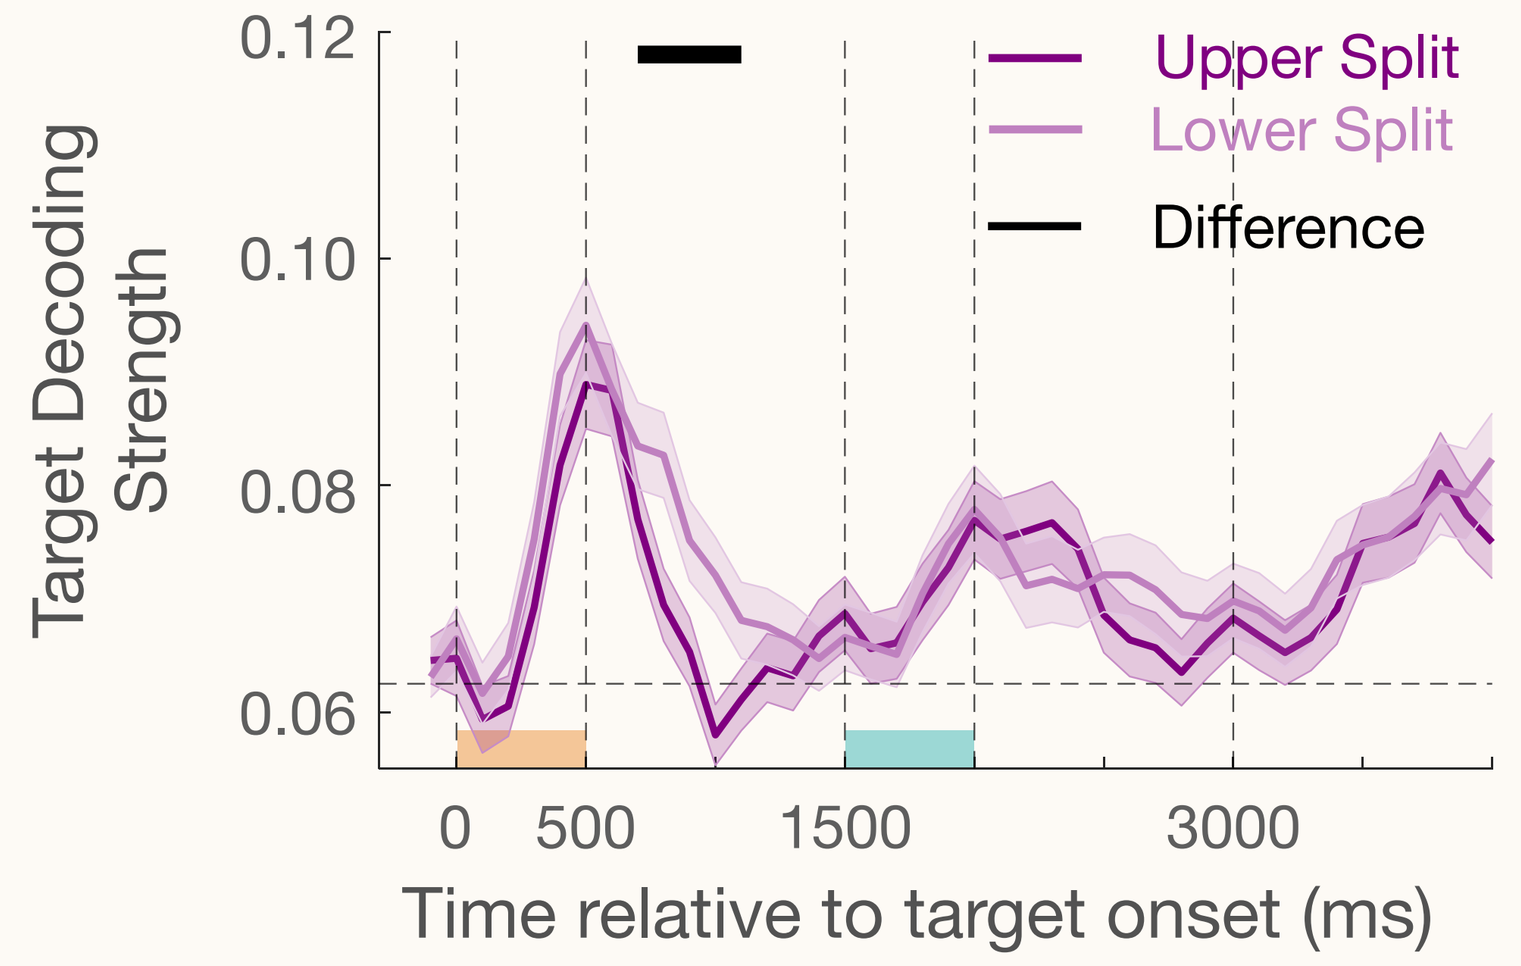

Target Decoding Strength
Upper Split
Lower Split
Difference
0.12
0.10
0.08
0.06
0
500
1500
3000
Time relative to target onset (ms)

Weaker target *(and potentially probe)* **decoding on trials with stronger SIMB!**

Is Probe Strength Related to Behavioral Offset?

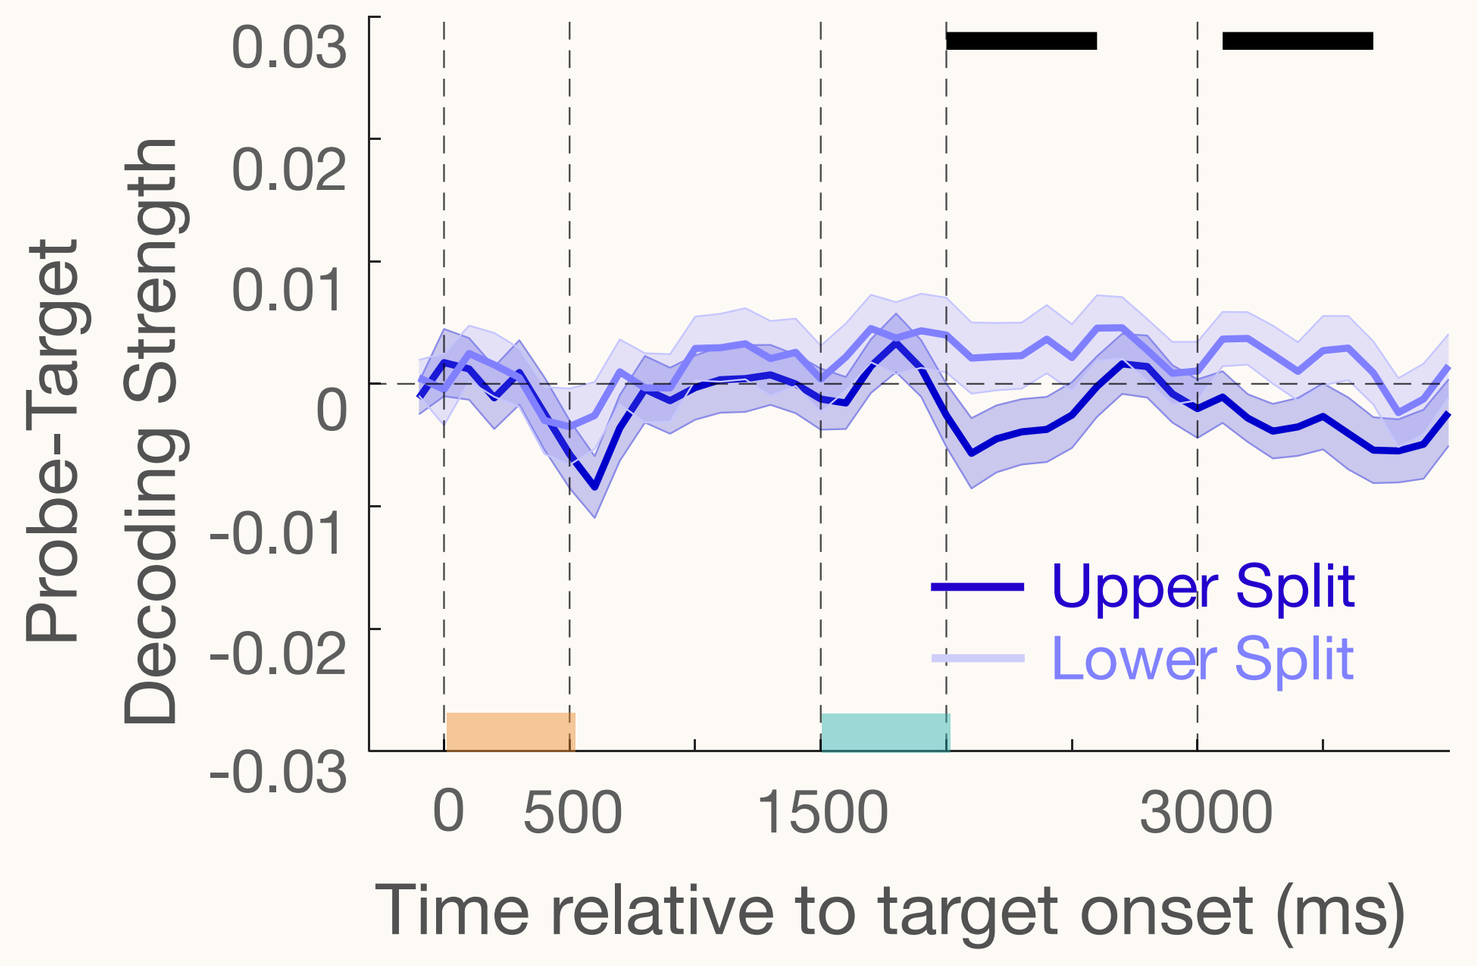

Probe-Target Decoding Strength
Upper Split
Lower Split
0.03
0.02
0.01
0
-0.01
-0.02
-0.03
0
500
1500
3000
Time relative to target onset (ms)

Counterintuitively, less probe decoding on trials with stronger bias toward probe.

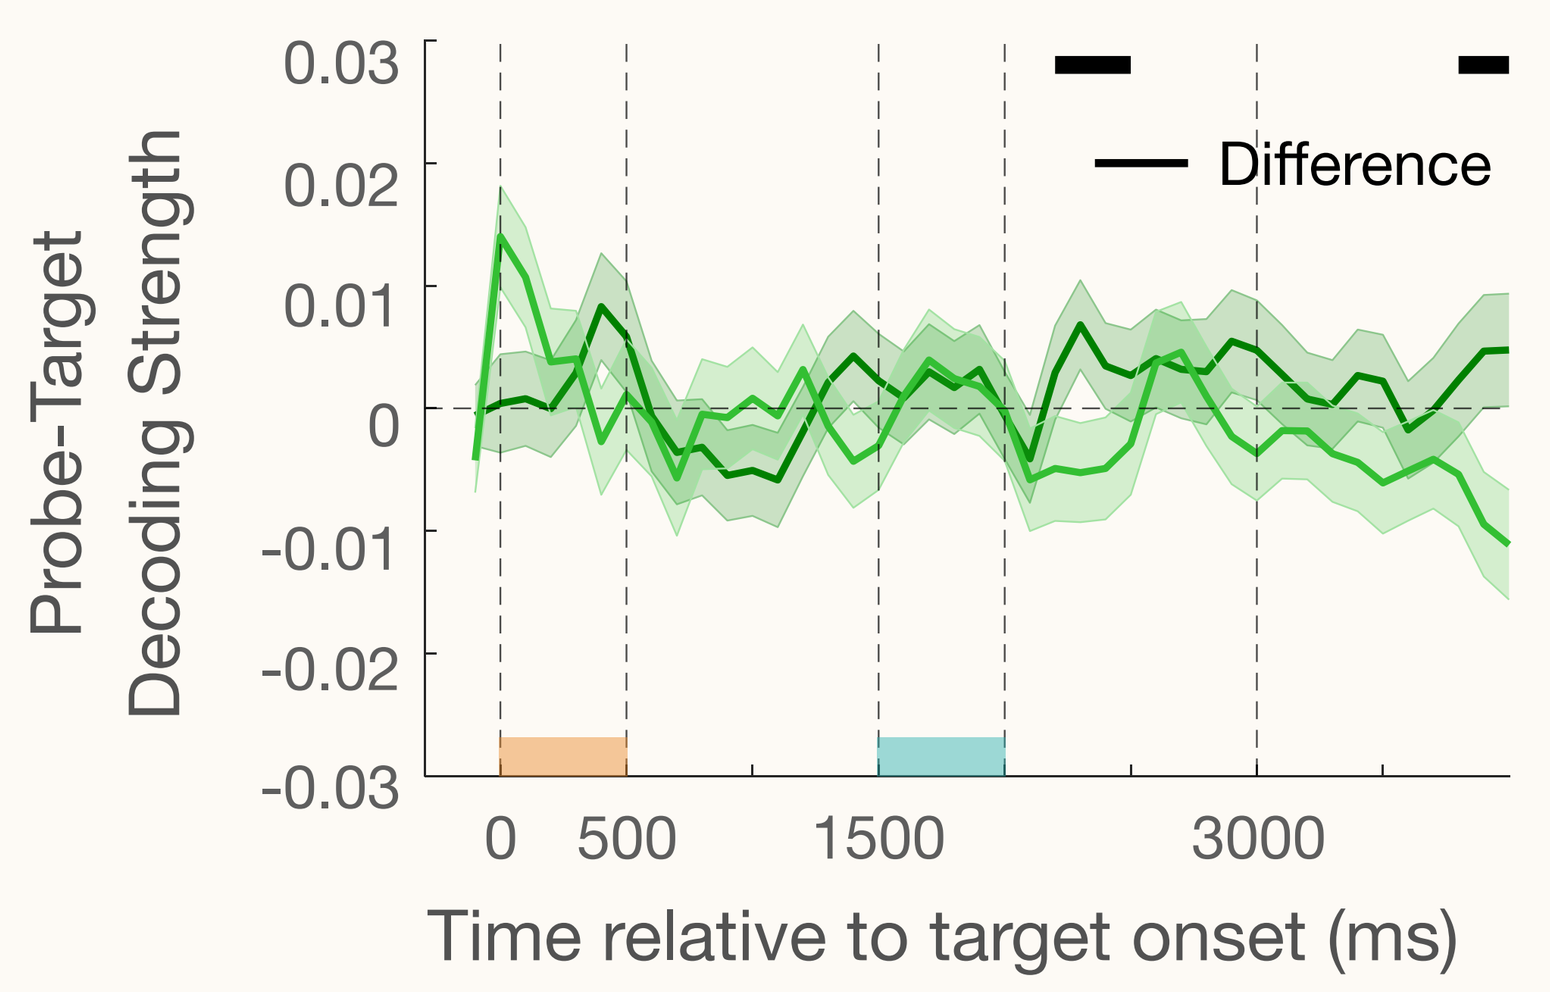

Probe-Target Decoding Strength
Difference
0.03
0.02
0.01
0
-0.01
-0.02
-0.03
0
500
1500
3000
Time relative to target onset (ms)

Stronger SIMB trials show stronger probe evidence after probe onset!

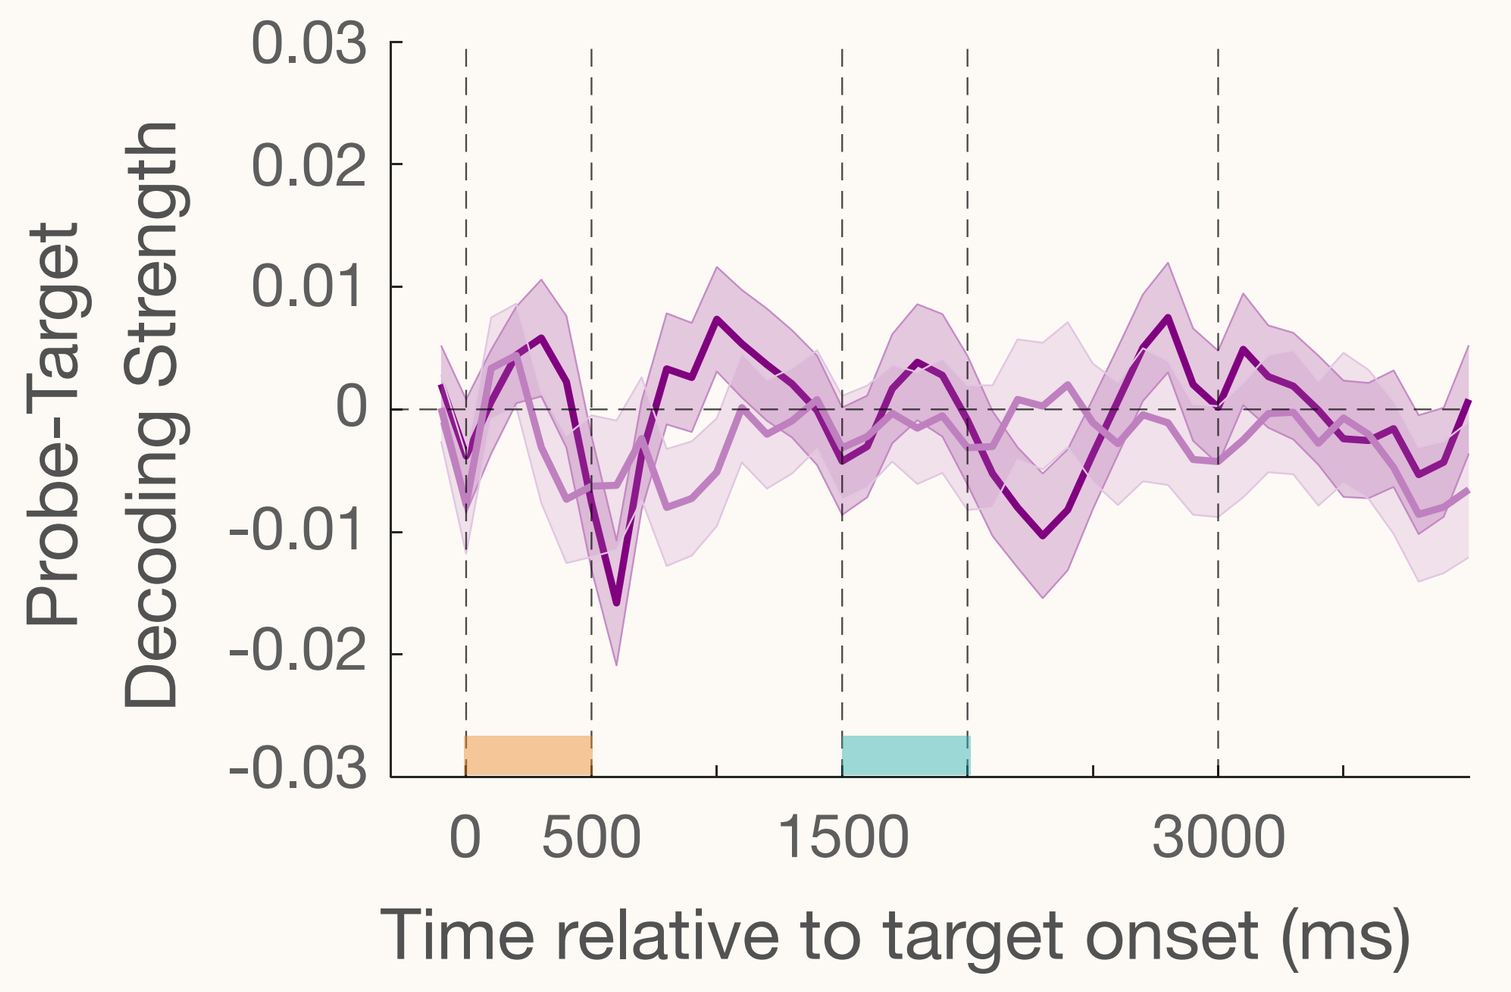

Probe-Target Decoding Strength
0.03
0.02
0.01
0
-0.01
-0.02
-0.03
0
500
1500
3000
Time relative to target onset (ms)

No difference between upper and lower split trials!

Conclusion

Weaker target encoding strength results in stronger SIMB.
Both encoding strength and probe interference modulate biases in VWM.

Reach Out!

Rademaker Lab

Fukudalab Lab

nursima.unver@mail.utoronto.ca

Random Thanks to Amit:

For being the amazing yo-yo master, winding me through science and life

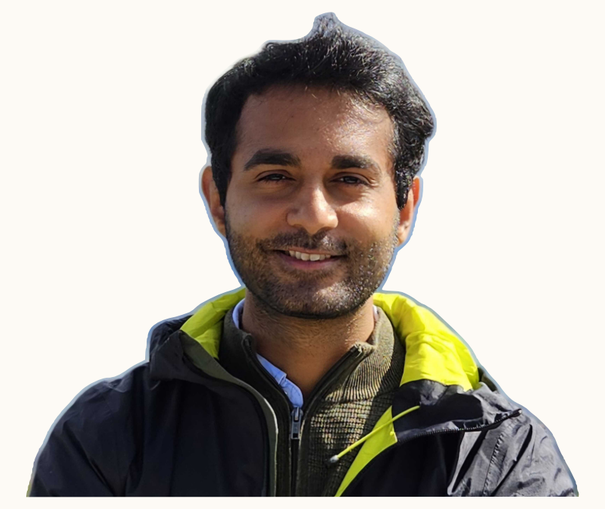

Check out his cool poster!! Right now, same room 56.426